Efficacy of Sublingual Apomorphine (APL-130277) for the Treatment of OFF Episodes in Patients with Parkinson's disease

Robert A. Hauser[1], MD; Jordan Dubow[2], MD; Bruce Dzyngel[2]; Thierry Bilbault[2], PhD; Anthony Giovinazzo[2]; Holly Shill[3], MD; Albert Agro[2], PhD

[1]University of South Florida Health Byrd Parkinson's Disease and Movement Disorders Center of Excellence, Tampa, FL, United States [2]Cynapsus Therapeutics, Toronto, Ontario, Canada [3]Banner Sun Health Research Institute, Sun City, AZ, United States

CYNAPSUS

Presented at the Movement Disorder Society's 19th Annual International Congress of Parkinson's Disease and Movement Disorders • San Diego, CA, United States • June 14–18, 2015

BACKGROUND

- Parkinson's disease (PD) patients suffer from a variety of OFF episodes as the disease progresses
- These consist of predictable wearing OFF, morning akinesia, delayed or No-ON or sudden OFF
- Up to 2/3rds of all PD patients across all stages of the disease experience OFF episodes, which have a significant negative impact on quality of life
- OFF time can be reduced by increasing the frequency of levodopa or by adding other adjunctive PD medications but, despite these manipulations, many PD patients suffer OFF episodes daily
- The only approved, acute treatment of OFF episodes is subcutaneous apomorphine (Apokyn® in the US), which is very efficacious but has significant limitations due to the parenteral nature of administration
- More convenient, on-demand, medications for the management of OFF episodes are needed
- APL-130277 is a soluble film strip of apomorphine
- It is administered sublingually to acutely manage OFF episodes by rapidly delivering apomorphine through absorption from the oral cavity mucosa
- APL-130277 is a "turning ON" medication
- This Phase 2 Study examined the effects of APL-130277 in PD patients with OFF episodes

OBJECTIVE

The primary objective of the study was to evaluate the efficacy, tolerability and safety of single treatments of APL-130277 in 19 PD patients with OFF episodes

METHODS

- This was a Phase 2, open-label, single-arm study
- Patients were instructed to take their last dose of levodopa no later than 10 PM the night prior and present to the clinic in the morning without taking their usual morning dose of levodopa and other PD medications
- Those patients confirmed to be in the OFF state were then dosed with APL-130277 (Figure 1), starting with 10 mg. If a full ON, as assessed by the Investigator and patient, was not achieved, the dose was increased in 5 mg increments until a full ON was achieved, to a maximum dose of 30 mg
- Patients could be dosed up to two times a day over 3 days
- If a patient achieved a full ON response, they received a subsequent confirmatory dose to verify the full ON response
- All patients were pretreated with trimethobenzamide for 3 days prior to initiation of APL-130277, which was continued during dosing
- Change in MDS-UPDRS Part III and assessment of OFF/ON were conducted pre-dose and at 15, 30, 45, 60 and 90 minutes after APL-130277 administration

Figure 1: APL-130277 sublingual apomorphine strip

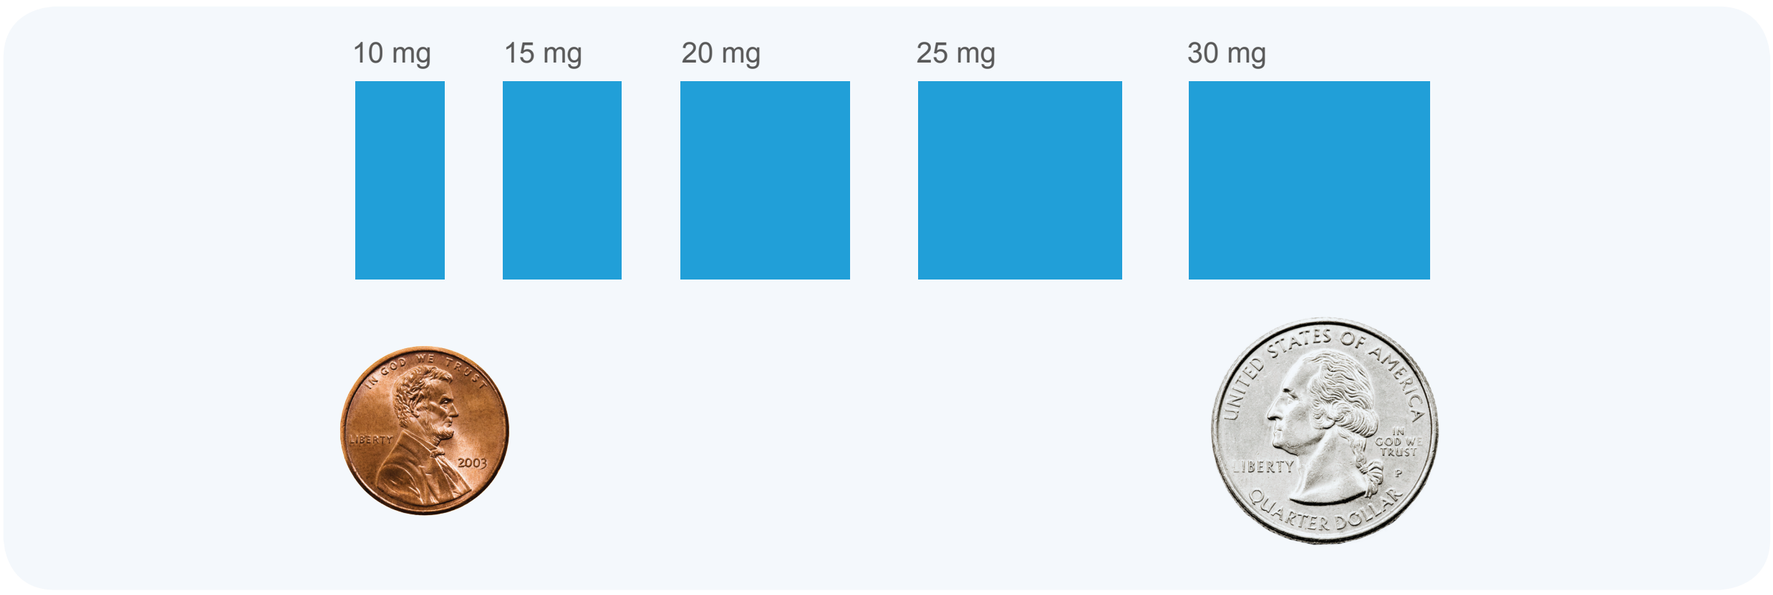
10 mg
15 mg
20 mg
25 mg
30 mg

Patients

- Main inclusion criteria
 - Clinical diagnosis of Parkinson's disease
 - At least one OFF episode per day and ≥ 2 hours of daily OFF time
 - Experiencing predictable OFF episodes upon awakening prior to receiving morning dose of levodopa
 - Hoehn and Yahr stage I–III in the ON state
- Main exclusion criteria
 - Atypical or secondary Parkinsonism
 - Past treatment with any form of apomorphine within 30 days of dosing Day 1

Efficacy & safety endpoints

- The primary efficacy endpoint was the percent of patients turning fully ON following an APL-130277 administration
- Secondary endpoints included the change and percent change in MDS-UPDRS Part III over time, percent of patients fully ON at each time point, and percent of patients with a 5 and 10 point MDS-UPDRS Part III improvement following the first full ON dose for Responders or last dose for non-responders
- Adverse events (AEs) were coded using the Medical Dictionary for Regulatory Activities (MedDRA) and were tabulated by MedDRA preferred term (PT).
- ECG, vital signs (including orthostatic blood pressure) and clinical laboratory values were evaluated
- Data were analyzed according to 3 datasets
 - Intention to Treat (ITT) – includes all 19 patients dosed
 - Responders – includes all 15 patients who turned fully ON following an APL-130277 administration
 - Per Protocol (PP) – includes 15 patients with no protocol dosing violations (excludes 3 patients who were instructed to swallow the strip instead of dissolve sublingually over 2 minutes and 1 patient who was dosed in an OFF state following administration of their first dose of PD medications)

RESULTS

- A total of 19 patients were dosed with APL-130277
- Baseline demographics are summarized in Table 1

Table 1: Baseline demographics

Mean age	61.5 (48-79)
Male: Female	14 (73.7%): 5 (26.3%)
Modified Hoehn & Yahr	2.2 (1-3)
Mean # of daily OFF episodes	3.9 (1-7)
Mean # PD medications classes	3 (1-5)
Mean daily levodopa dose (mg)	837 (100-1500)
Mean # levodopa doses per day	5.3 (1-12)

- Of the 19 total patients dosed with APL-130277, 15 achieved a full ON response (Figure 2)
 - 6 patients turned fully ON within 15 minutes
 - 9 patients turned fully ON between 15 and 30 minutes
 - 13/15 remained fully ON for at least 30 minutes
 - 9/15 remained fully ON for at least 60 minutes
 - Mean ON duration was 52 minutes
 - One subject who was dosed incorrectly (told to swallow instead of dissolve sublingually) turned ON but only had a duration of ON of 15 minutes
 - Of the 4 non-responders, 2 were dosed incorrectly (told to swallow the strip instead of dissolve sublingually at all doses 10–30 mg) and 2 were dosed up to the maximum dose of 30 mg

Figure 2: Percent of patients fully ON overall and at each time-point following APL-130277 administration

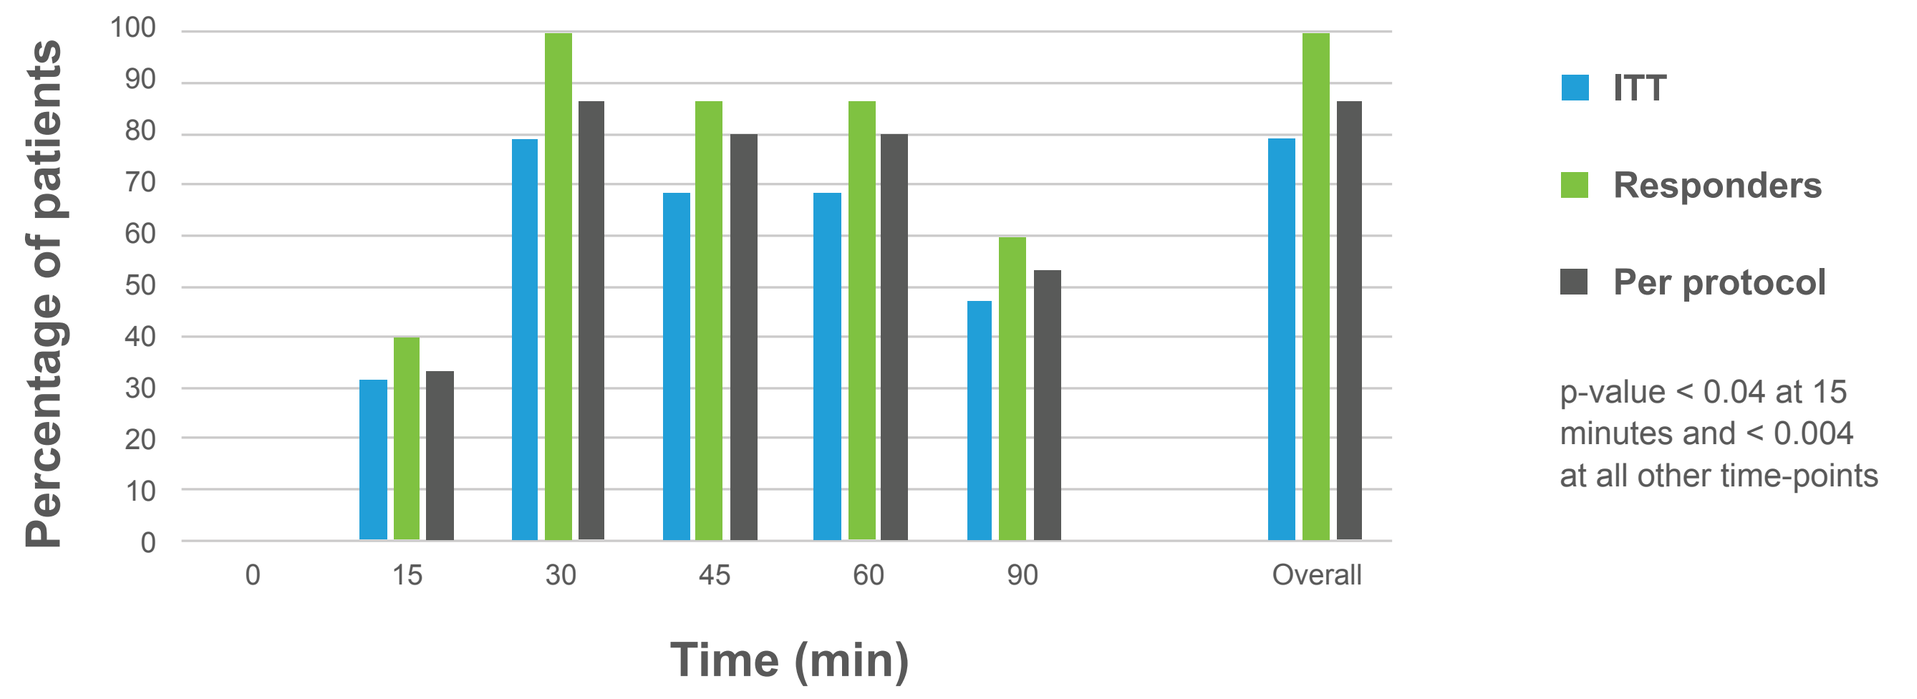
Percentage of patients
Time (min)
0
15
30
45
60
90
Overall
ITT
Responders
Per protocol
p-value < 0.04 at 15 minutes and < 0.004 at all other time-points

- 80% of patients turned fully ON with 20 mg or less and over half with 15 mg or less (Figure 3)

Figure 3: APL-130277 dose distribution at first full ON

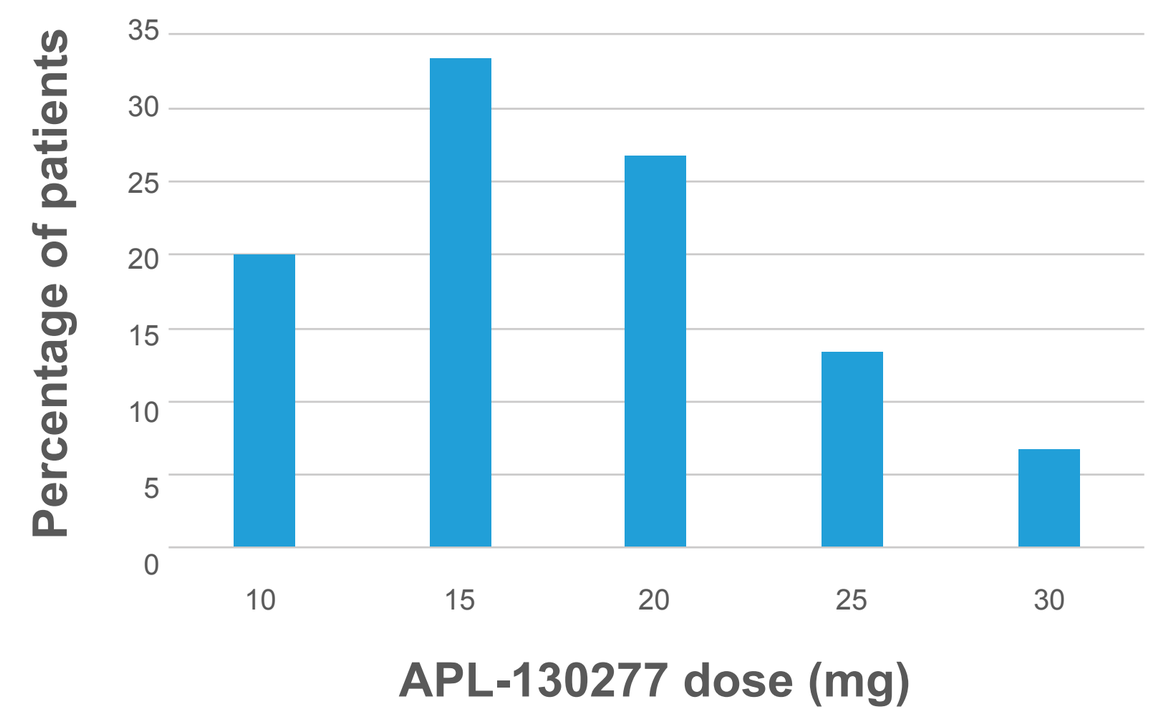
Percentage of patients
APL-130277 dose (mg)
10
15
20
25
30

- Mean change and percent change in MDS-UPDRS Part III for the first full ON for Responders or last dose for non-responders is presented in Figures 4 and 5 and Table 2
 - A large, robust, statistically significant and clinically meaningful improvement in motor function was seen at all-time points
 - Maximum change at any time-point was -18.9 for ITT and -20.5 for responders
 - An approximately 30% or greater MDS-UPDRS Part III improvement was seen at all time-points with a maximum mean percent change at any time-point of -45.6 for ITT and -51.4 for responders
 - Confirmatory dosing revealed a similar improvement in the MDS-UPDRS Part III

Figure 4: Mean MDS-UPDRS part III change over time following APL-130277 administration

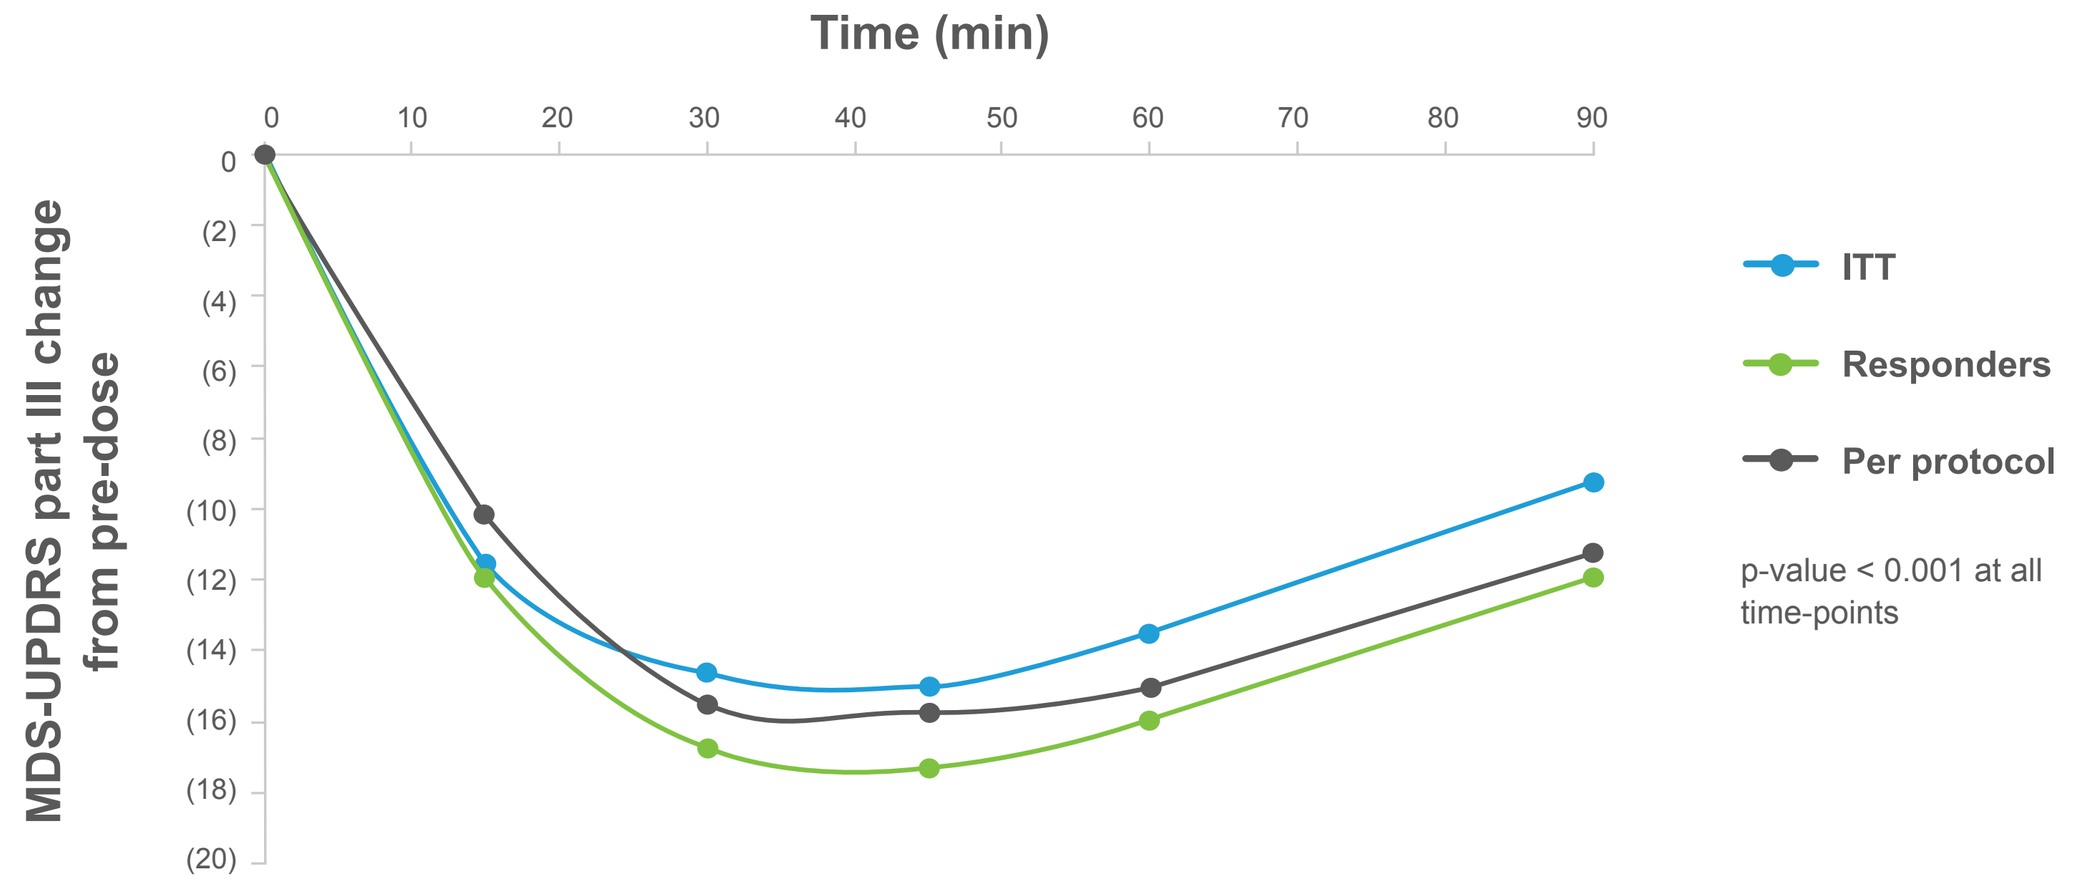
Time (min)
MDS-UPDRS part III change from pre-dose
ITT
Responders
Per protocol
p-value < 0.001 at all time-points

Table 2: Mean change (standard deviation) in MDS-UPDRS part III over time

Time (min)	15	30	45	60	90
ITT	-11.5 (9.5)	-14.6 (9.1)	-15.0 (9.5)	-13.5 (9.7)	-9.2 (10.0)
Responders	-11.9 (9.5)	-16.7 (8.9)	-17.3 (9.1)	-15.9 (9.5)	-11.9 (9.5)
Per protocol	-10.1 (10.0)	-15.5 (9.6)	-15.7 (9.4)	-15 (8.6)	-11.2 (9.8)

Figure 5: Mean percent change in MDS-UPDRS part III following APL-130277 administration

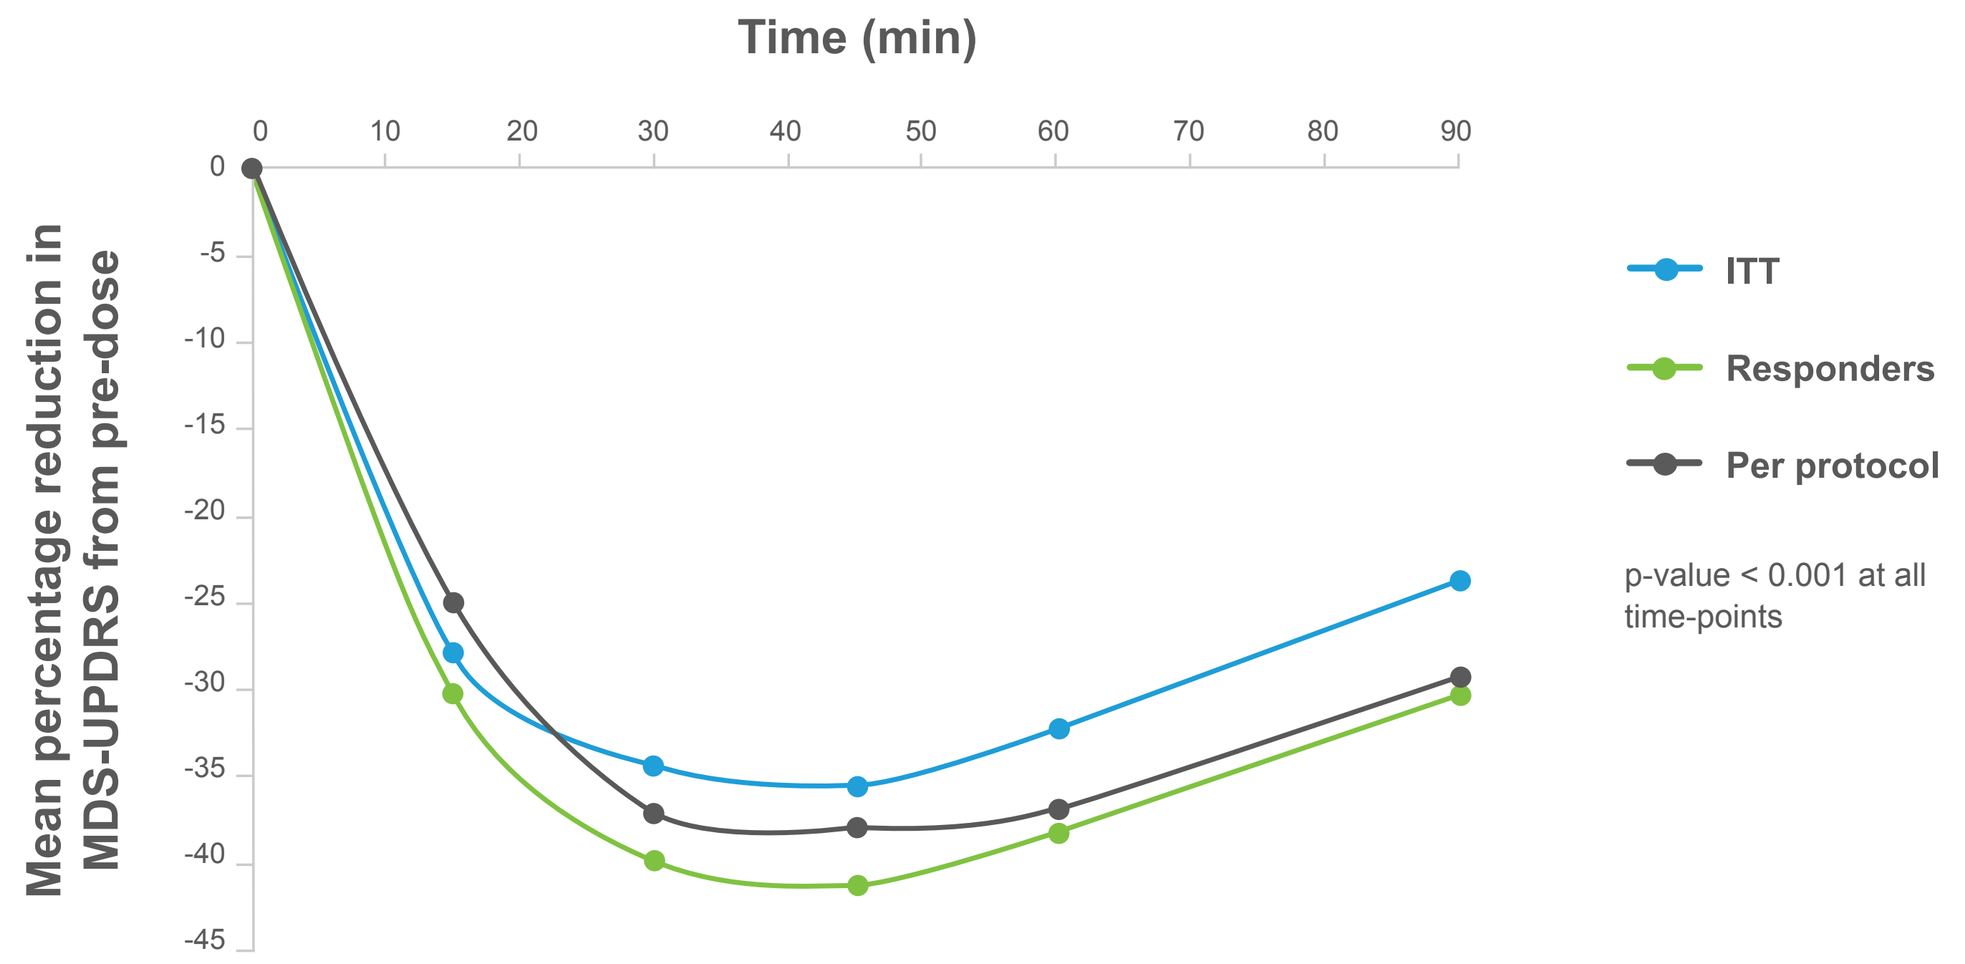
Time (min)
Mean percentage reduction in MDS-UPDRS from pre-dose
ITT
Responders
Per protocol
p-value < 0.001 at all time-points

- A large percentage of patients had a 5 or 10 point improvement in the MDS-UPDRS Part III overall and at each time-point (Figure 6 and 7)
 - 90% of patients in the ITT had a 5 point or greater improvement at any time-point
 - 85% of patients in the ITT had a 10 point or greater improvement at any time-point

Figure 6: Percentage of patients with a 5 point or greater MDS-UPDRS Part III improvement by time-point

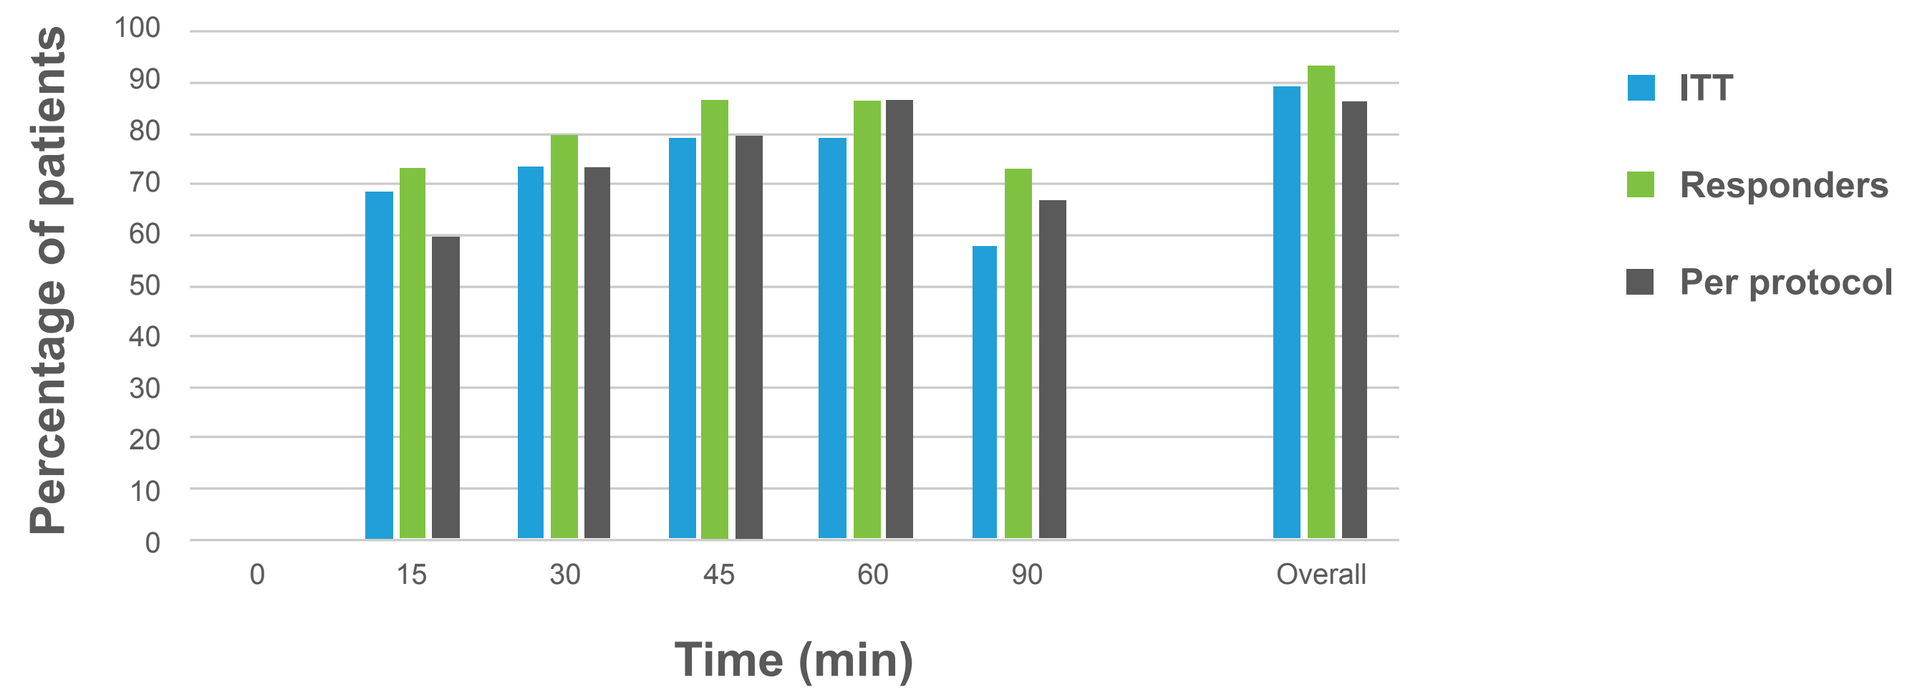
Percentage of patients
Time (min)
Overall
ITT
Responders
Per protocol

Figure 7: Percentage of patients with a 10 point or greater MDS-UPDRS Part III improvement by time-point

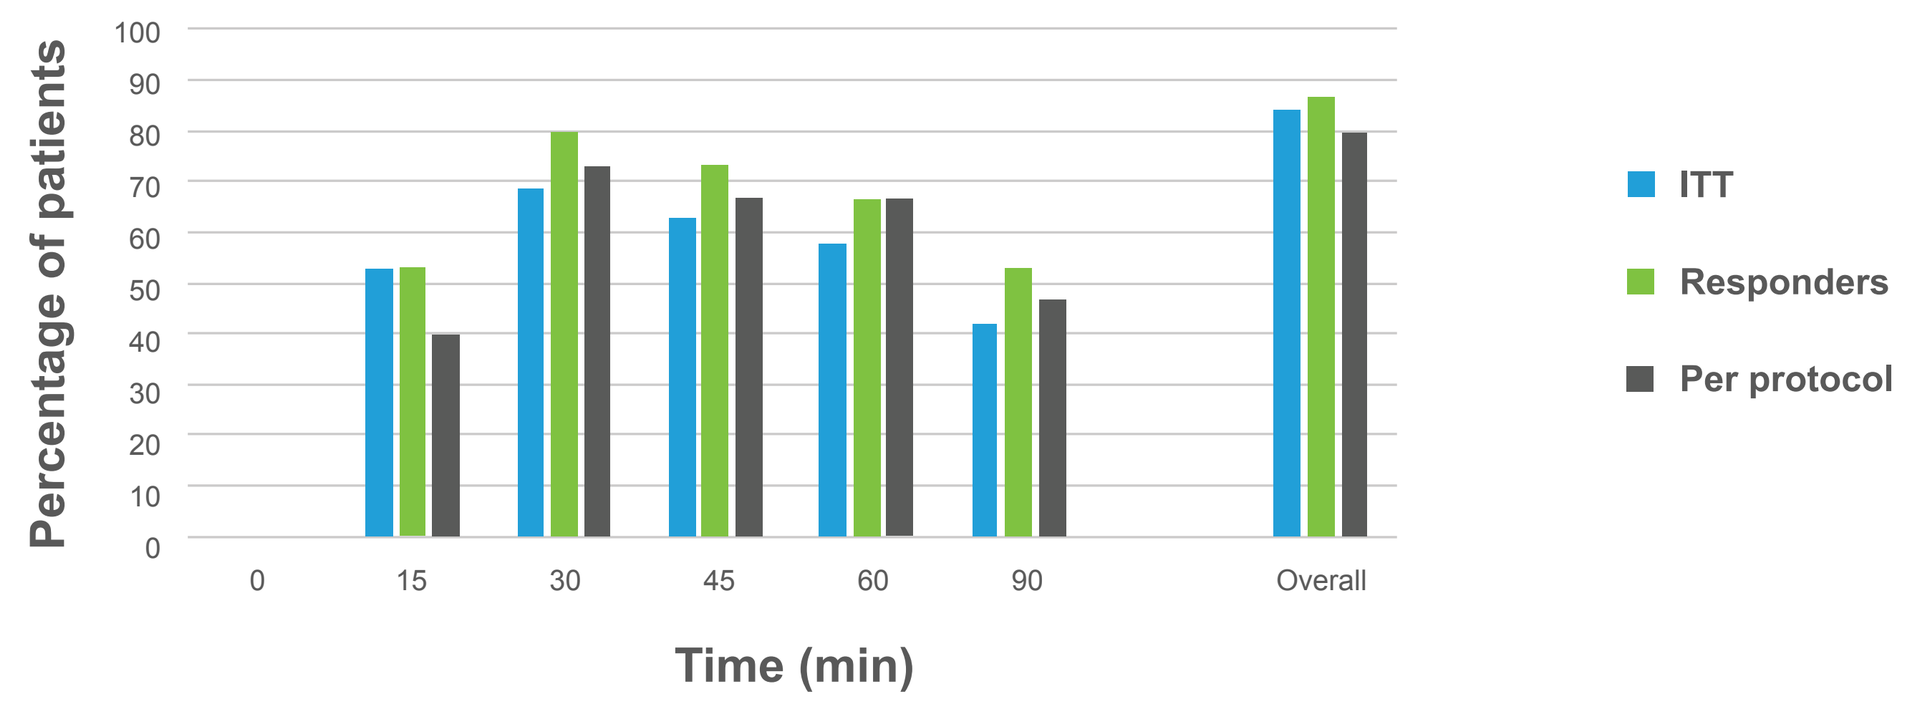
Percentage of patients
Time (min)
Overall
ITT
Responders
Per protocol

Safety

- Full Safety Data is presented as a poster presentation at the 19th International Congress of Parkinson's Disease and Movement Disorders (abstract # 243)
- Overall, APL-130277 was safe and well tolerated
- Adverse events (AEs) seen were known dopaminergic adverse events and were mostly mild or moderate
- The was one unrelated serious AE of dysphagia
- There were no discontinuations due to AEs
- There were no signs of local mucosal irritation

CONCLUSIONS

- Sublingual APL-130277 rapidly converted PD patients from the morning OFF state to the ON state
- APL-130277 provided, rapid, clinically meaningful improvement in motor function as assessed by MDS-UPDRS Part III scores
- Duration of benefit was close to an hour on average with most patients having sustained benefit through 90 minutes after APL-130277 administration
- A range of doses were utilized but over half of patients responded to the two lowest doses
- APL-130277 was generally well tolerated
- APL-130277 may be an effective, easy to administer medication for the acute, on-demand management of OFF episodes in PD patients
- Phase 3 studies are being initiated to further evaluate the efficacy, safety, tolerability of APL-130277 in PD patients

REFERENCES

1. Aquino C, Fox S. Clinical spectrum of levodopa-induced complications. Movement Disorders. 2015;30: 80-89.
2. Rizos A, Martinez-martin P, Odin P, et al. Characterizing motor and non-motor aspects of early-morning OFF periods in Parkinson's disease: an international multicenter study. Parkinsonism and Related Disorders. 2014;20:1231-1235.
3. Chapuis S, Ouchchane L, Metz O, et al. Impact of motor complications of Parkinson's disease on the quality of life. Movement Disorders. 2005;20:224-230.
4. Apokyn USPI, US WorldMeds, LLC. Louisville, KY, 2014.
5. Laar T, van der Geest R, Danhof M, et al. Stepwise intravenous infusion of apomorphine to determine the therapeutic window in patients with Parkinson's disease. Clinical Neuropharmacology. 1998;21:152-158.

ACKNOWLEDGEMENTS

This study was supported by Cynapsus Therapeutics. Additionally, the Michael J. Fox Foundation provided a grant in support of the study. JD, BD, TB, AG and AA are all employees of Cynapsus Therapeutics and hold stock or stock options. RAH and HS were Investigators in the study and received compensation from Cynapsus for research activities. Peter Gardzinski, Emily Calaiezzi, Gazal Vakili and Lewis Lau of Cynapsus Therapeutics provided assistance in the development of this publication.
APL-130277 is currently an investigational product in some countries, including the United States.

THE MICHAEL J. FOX FOUNDATION FOR PARKINSON'S RESEARCH

cynapsus.ca